Poster Number

494

11th CROI
February 8-12, 2004
San Francisco, California

Broad-Spectrum Micronutrient Supplementation in HIV-infected Patients With Dideoxynucleoside-related Peripheral Neuropathy: A Prospective, Double-Blind, Placebo Controlled Trial

J Kaiser*[1], J Ondercin[2], G Santos[3], G Leoung[4], S Brown[5], M Mass[6], M Baum[7]

[1]University of California at San Francisco, SF, CA; [2]Jonathan Lax Treatment Center; Philadelphia, PA; [3]Betances Health Center, NYC, NY;
[4]HIVCare, Saint Francis Memorial Hospital, SF, CA; [5]AIDS ReSearch Alliance, West Hollywood, CA;
[6]California Pacific Medical Center, SF, CA; [7]Florida International University, Miami, FL

Integrative Health Consulting, Inc.
655 Redwood Highway, Suite 225
Mill Valley, CA 94941
Tel: (415) 381-7505
Fax: (415) 381-7503
www.integrativehealthconsulting.com

This micronutrient formula, as well as its matching placebo, can be obtained by emailing ihcmd7@mac.com.

Objectives

- A prospective, randomized, double blind, placebo-controlled trial to study the effects of high-dose micronutrients in HIV-infected patients with drug-induced peripheral neuropathy on stable HAART
- Primary and Secondary Endpoints included:
 a. Effect on Neuropathy Inventory Linear Analog Scale (NILAS)
 b. Effect on quality of life utilizing Linear Analog Scale Assessment (LASA) and MOS-HIV assessment tools
 c. Effect on CD4 count and HIV RNA viral load
 d. Effect on Fasting Lipids, Glucose, Serum Lactate, and ALT levels
 e. Overall Tolerability of the High-Dose Micronutrient Composition

Rationale

Peripheral neuropathy (PN) occurs in upwards of 10% of patients beginning antiretroviral therapy with either stavudine (D4T) or didanosine (DDI)- containing antiviral regimens.[1]

Stavudine and didanosine have been shown to significantly inhibit the production of mitochondrial DNA in peripheral neurons. Therefore, a buildup of free oxygen species (FOS) toxicity might eventually lead to the occurrence of mitochondrial dysfunction. This mechanism has been proposed as a cause of dideoxynucleoside-induced peripheral neuropathy.[2]

Healthy mitochondrial functioning is a highly nutrient dependent process. Acetyl L-carnitine is utilized to transport free fatty acids across the mitochondrial membrane to be used as fuel. N-acetyl-cysteine (NAC) and alpha-lipoic acid are key molecules which are used to neutralize the oxidation byproducts of energy metabolism (i.e. free radicals). Vitamins C, E, and selenium are also utilized in the neutralization of cytotoxic free radicals.

If any of these nutrient-dependent pathways becomes depleted, increased mitochondrial toxicity leading to cell death (apoptosis) is more likely to occur.[3]

Based on the above information, we tested a potent antioxidant formula designed to lessen the systemic clinical effects of mitochondrial toxicity, including the symptoms of PN, in HIV-infected individuals taking a stable, D-drug-containing HAART regimen. Laboratory measurements were used to assess the metabolic and anti-HIV effects of this high-dose micronutrient formula.

Methods

- **Forty (40) patients with symptomatic peripheral neuropathy taking either stavudine or didanosine** were randomly assigned to take a high-dose micronutrient or placebo packet twice daily during 12 weeks in double blinded fashion at four research centers in the US.
- **All study subjects were required to be on a stable HAART regimen** and not to consume any other micronutrients in excess of one low-dose multivitamin pill per day.
- **The micronutrient compound tested included substantial dosages of 3 potent antioxidants:** acetyl L-carnitine (1,000 mg/day), n-acetyl cysteine (1,200 mg/day), and alpha lipoic acid (400 mg/day) mixed with a high potency multivitamin/ multimineral formula (Table 1).
- **The manufacture of the high-dose micronutrients did not include** any fillers, binders, and/or lubricants (e.g. stearates or palmitates) which are commonly used in commercial supplement manufacturing. These substances can inhibit the absorption, bioavailability, and/or tolerance of nutritional supplements in metabolically compromised individuals.
- **Data was collected at 4-week intervals and consisted of:**
 1. Neuropathy linear analog scale (NILAS)
 2. Quality of life measurements (LASA and MOS-HIV)
 3. CD4 count and HIV RNA (by PCR)
 4. Serum lactate, ALT; fasting glucose, insulin, and lipids
 5. A focused neurological exam (NEAT)
- **Statistical analyses** were executed using two sided t-tests with an alpha level of 0.05. No adjustments were made for multiple comparisons. Missing values were imputed using a last-observation-carried-forward method.

Table 1. Broad-Spectrum Micronutrient Formula (Total Daily Dosage)

Multivitamin		Multimineral	
Beta Carotene	20,000 i.u.	Calcium	800 mg
Vitamin C	1,800 mg	Iron	18 mg
Vitamin D	400 i.u.	Iodine	150 mcg
Vitamin E	800 i.u.	Magnesium	400 mg
Vitamin B1	60 mg	Zinc	30 mg
Vitamin B2	60 mg	Selenium	200 mcg
Vitamin B6	260 mg	Copper	2.0 mg
Niacinamide	60 mg	Manganese	10 mg
Folic acid	800 mcg	Chromium	100 mcg
Vitamin B12	2.5 mg	Molybdenum	300 mcg
Biotin	50 mcg	Choline	60 mg
Inositol	60 mg	Glutamic acid	100 mg
Pantothenic acid	60 mg	Boron	2.0 mg
Potassium	99 mg	Betaine HCL	150 mg
Vitamin A	8,000 i.u.	Bioflavinoid complex	300 mg

Highly Potent Antioxidants	
Alpha lipoic Acid	400 mg
N-acetal cysteine (NAC)	1200 mg
Acetyl L-carnitine	1000 mg

Results

Table 2. Baseline Demographics and Clinical Characteristics

Variable	Placebo (n=22)	Micronutrients (n=18)
Mean age	46.6	46.5
Viral load (log10)	2.4	2.4
Mean CD4 count (cells)	467	356
Mean duration of neuropathy (in months)	12.2	21.4
On Stavudine (not taking didanosine)	54%	66%
On Didanosine (not taking stavudine)	23%	17%
On both Stavudine and Didanosine	23%	17%
Neuropathy Score (0 to 100) (100 = severe neuropathy symptoms)	57	52
QOL Score (0 to 100) (100 = ideal energy & QOL)	42	48

The treatment and placebo groups were similar at baseline in most respects. However, the mean duration of neuropathy symptoms was higher, and the CD4+ count lower, in the treatment group.

Table 3. Neuropathy and Quality of Life Changes (On Treatment n=28)

% Improvement	Placebo	Nutrients
Neuropathy Score (NILAS)	33%	**42%** (p=0.8)
Quality of Life Score (LASA)	28%	**29%** (p=0.8)

Patients taking the micronutrients showed a 42% improvement in neuropathy linear pain scale (NILAS) and a 29% improvement in quality of life score (LASA) over twelve weeks. A statistically significant difference from placebo was not evident.

The following figures are from the Intent to Treat Cohort (n=40)

MEAN PERCENT CHANGE CD4 COUNT

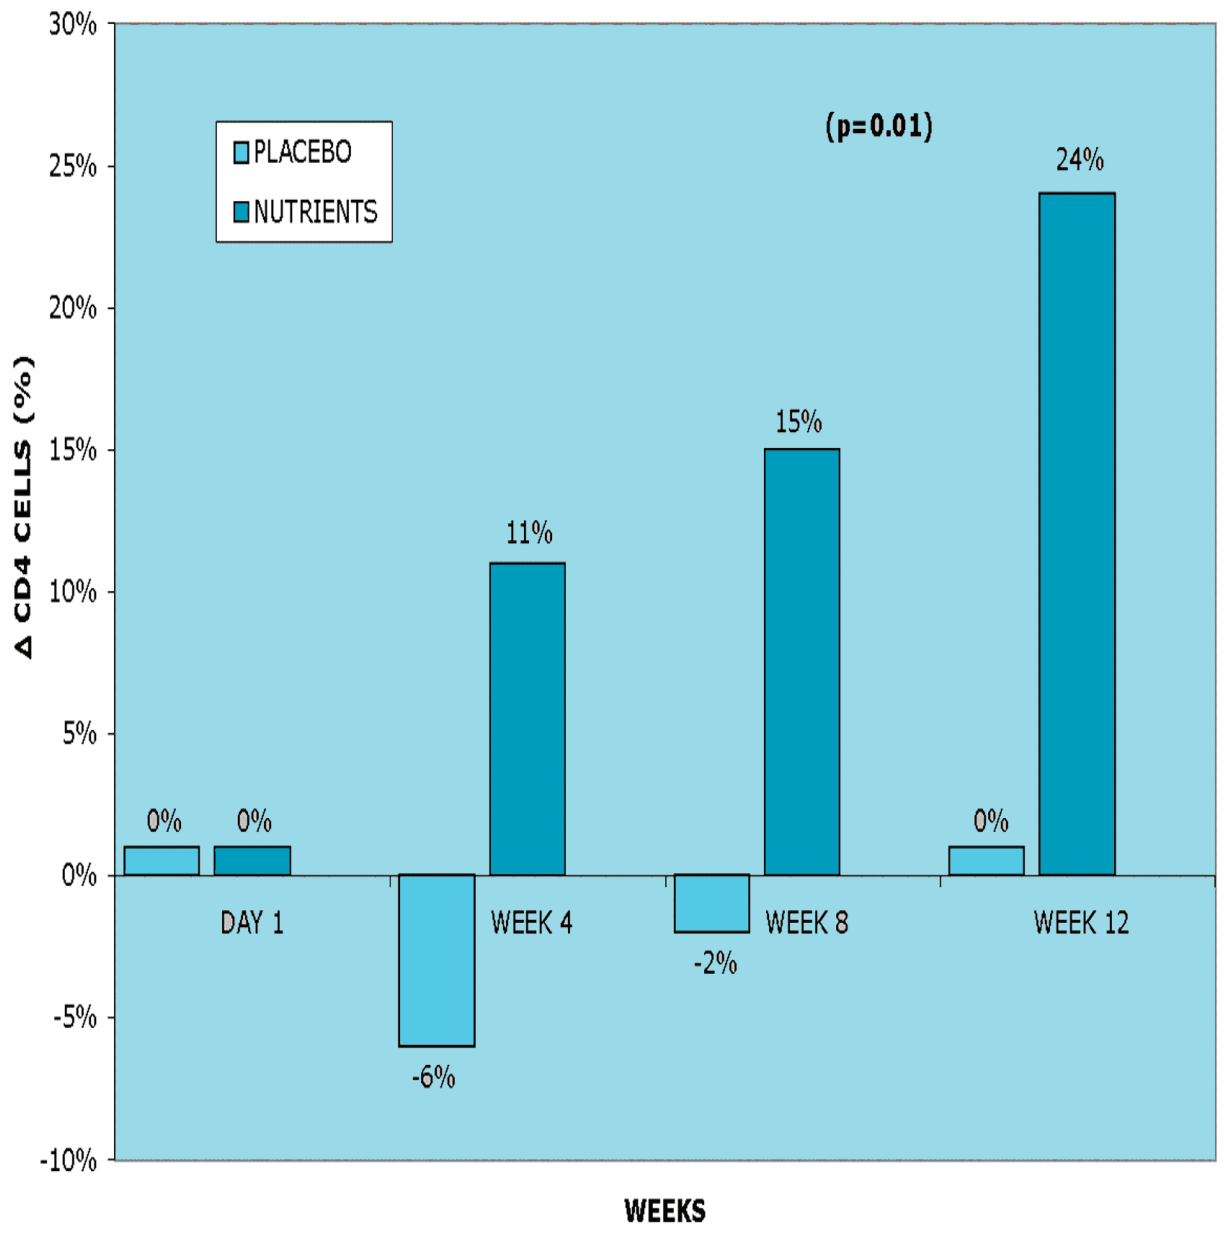

30%
25%
20%
15%
10%
5%
0%
-5%
-10%
Δ CD4 CELLS (%)
PLACEBO
NUTRIENTS
(p=0.01)
0%
0%
DAY 1
-6%
11%
WEEK 4
-2%
15%
WEEK 8
0%
24%
WEEK 12
WEEKS

FIGURE 1
Patients taking the micronutrients had an average increase of 24% in their CD4+ cell counts from pretreatment values compared to 0% in those taking a placebo. This difference was statistically significant (p=0.01 ITT).

MEAN CHANGE IN CD4 COUNT

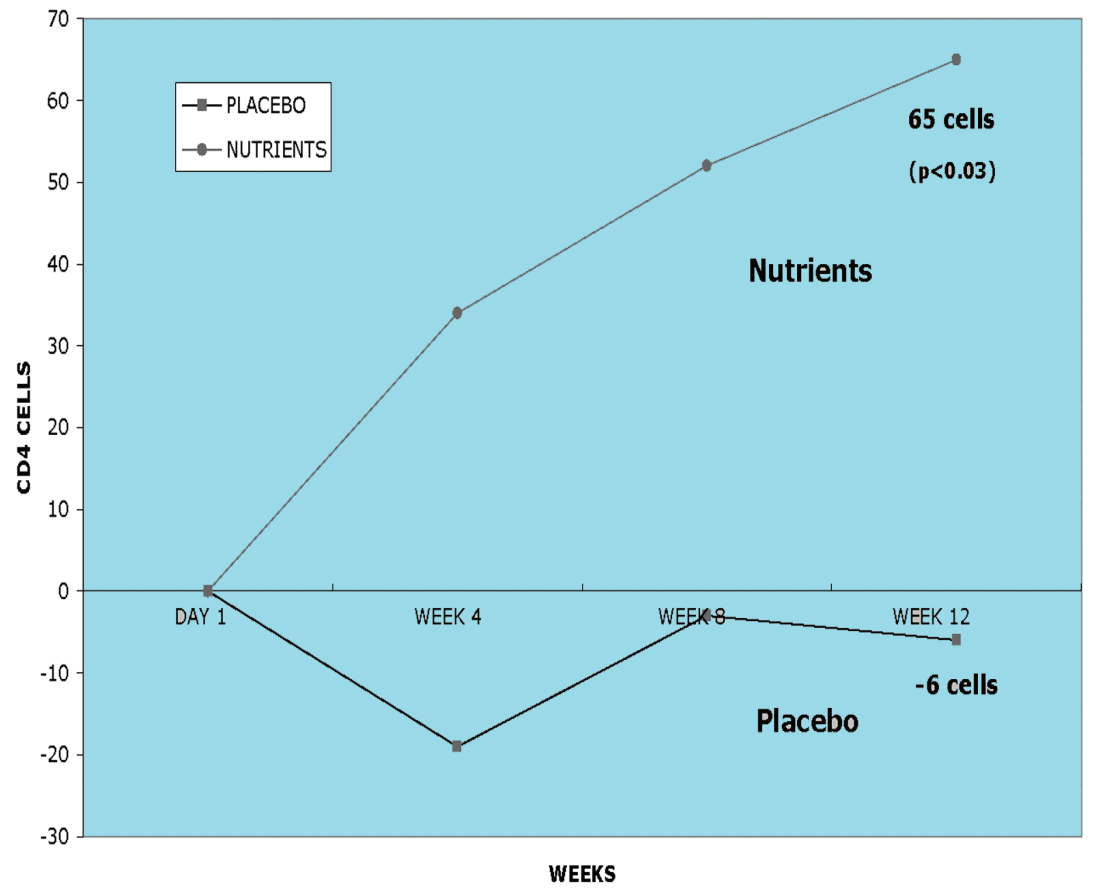

70
60
50
40
30
20
10
0
-10
-20
-30
CD4 CELLS
PLACEBO
NUTRIENTS
65 cells
(p<0.03)
Nutrients
DAY 1
WEEK 4
WEEK 8
WEEK 12
-6 cells
Placebo
WEEKS

FIGURE 2
The patients taking the micronutrients experienced a mean 65 cell rise in their absolute CD4+ cell count compared to a 6 cell decline in the patients taking the placebo. This difference was statistically significant (p<0.03 ITT).

MEAN Δ IN HIV RNA

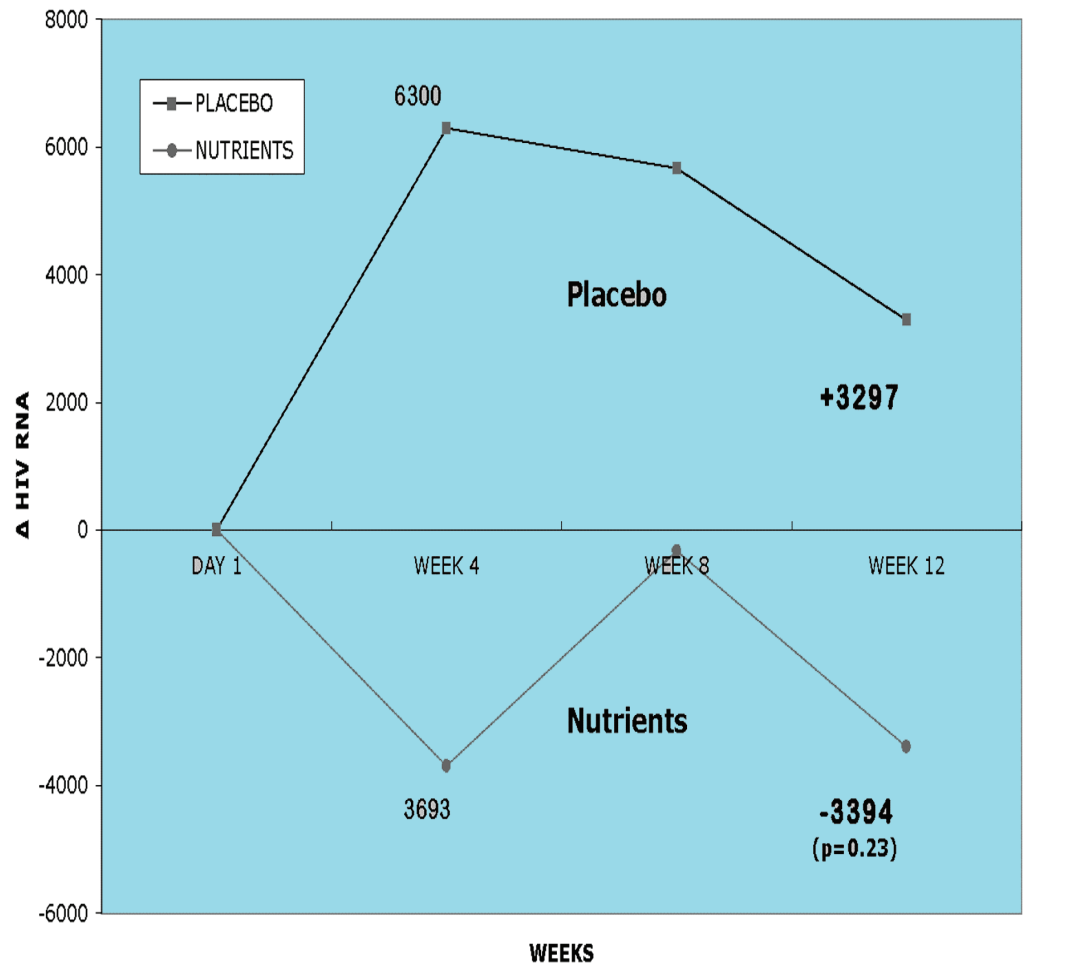

8000
6000
4000
2000
0
-2000
-4000
-6000
Δ HIV RNA
PLACEBO
NUTRIENTS
6300
Placebo
+3297
DAY 1
WEEK 4
WEEK 8
WEEK 12
Nutrients
3693
-3394
(p=0.23)
WEEKS

FIGURE 3
Patients taking the micronutrients experienced a mean decline of 3,394 copies in HIV RNA viral load compared to an increase of 3,297 copies in HIV RNA viral load in patients taking the placebo (p=0.23 ITT).

Metabolic Parameters

The effects of the nutrient composition on fasting glucose, insulin, cholesterol, triglycerides, serum lactate, and ALT levels were also assessed. The data collected indicates that none of these parameters were adversely affected in the patients taking the high-dose micronutrient supplements.

Adverse Events

There were two adverse events reported and both occurred in the treatment group (bacterial pneumonia and cystic lithiasis). Both were judged unrelated to the micronutrient treatment and both patients recovered uneventfully and completed the study. Of note, there were no reported GI side effects in either the placebo or high-dose micronutrient groups.

Conclusions

- HIV-infected individuals with long standing PN showed substantial improvement in both linear pain scale (42%) and quality of life measurements (29%) during twelve weeks of taking this micronutrient formula. A statistically significant difference from placebo was not shown.
- The high-dose micronutrient group showed a steady and statistically significant 26% mean percent rise in CD4 count over twelve weeks.
- The high-dose micronutrient group showed a steady and statistically significant 65 cell increase in CD4 count over twelve weeks.
- The high-dose micronutrient group showed a trend toward decreasing HIV RNA over twelve weeks of taking the micronutrient formula.
- The use of this broad-spectrum micronutrient formula may hold promise as a complementary therapy to HAART and should be further investigated.

References

1. Simpson DM, et al. Neurologic manifestations of HIV infection. *Ann Intern Med*. 1994;121:769-785
2. Chen C, et al. Effect of anti-human immunodeficiency virus nucleoside analogs on mitochondrial DNA and its implications for delayed toxicity. *Mol Pharmacol*. 1991;39:625-628
3. Butte TM, et al. Oxidative stress as a mediator of apoptosis. *Immunol Today* 1994; 15:7-10